ABSTRACT # 1213

ANG4043, a brain-penetrant anti-HER2 mAb, increases survival in mice bearing intracranial BT-474 breast tumor cells.

Betty Lawrence, Jean E. Lachowicz, Michel Demeule, Anthony Régina, Emilie Houle, Sasmita Tripathy, & Jean-Paul Castaigne.

Angiochem Inc., 201 President Kennedy Avenue, PK-2880, Montréal, QC, Canada H2X 3Y7

Angiochem BEYOND BARRIERS

INTRODUCTION

HER2+ Brain Metastases

- A critical unmet medical need
- In US and Europe, 465,000 new diagnoses of breast cancer per year
- Breast cancers overexpressing HER2+ represent ~116,000 patients
- About 50% of these will die from brain metastasis (55,000 pts)
 - Herceptin does not cross the BBB and therefore can only address peripheral metastases

Angiopep Technology

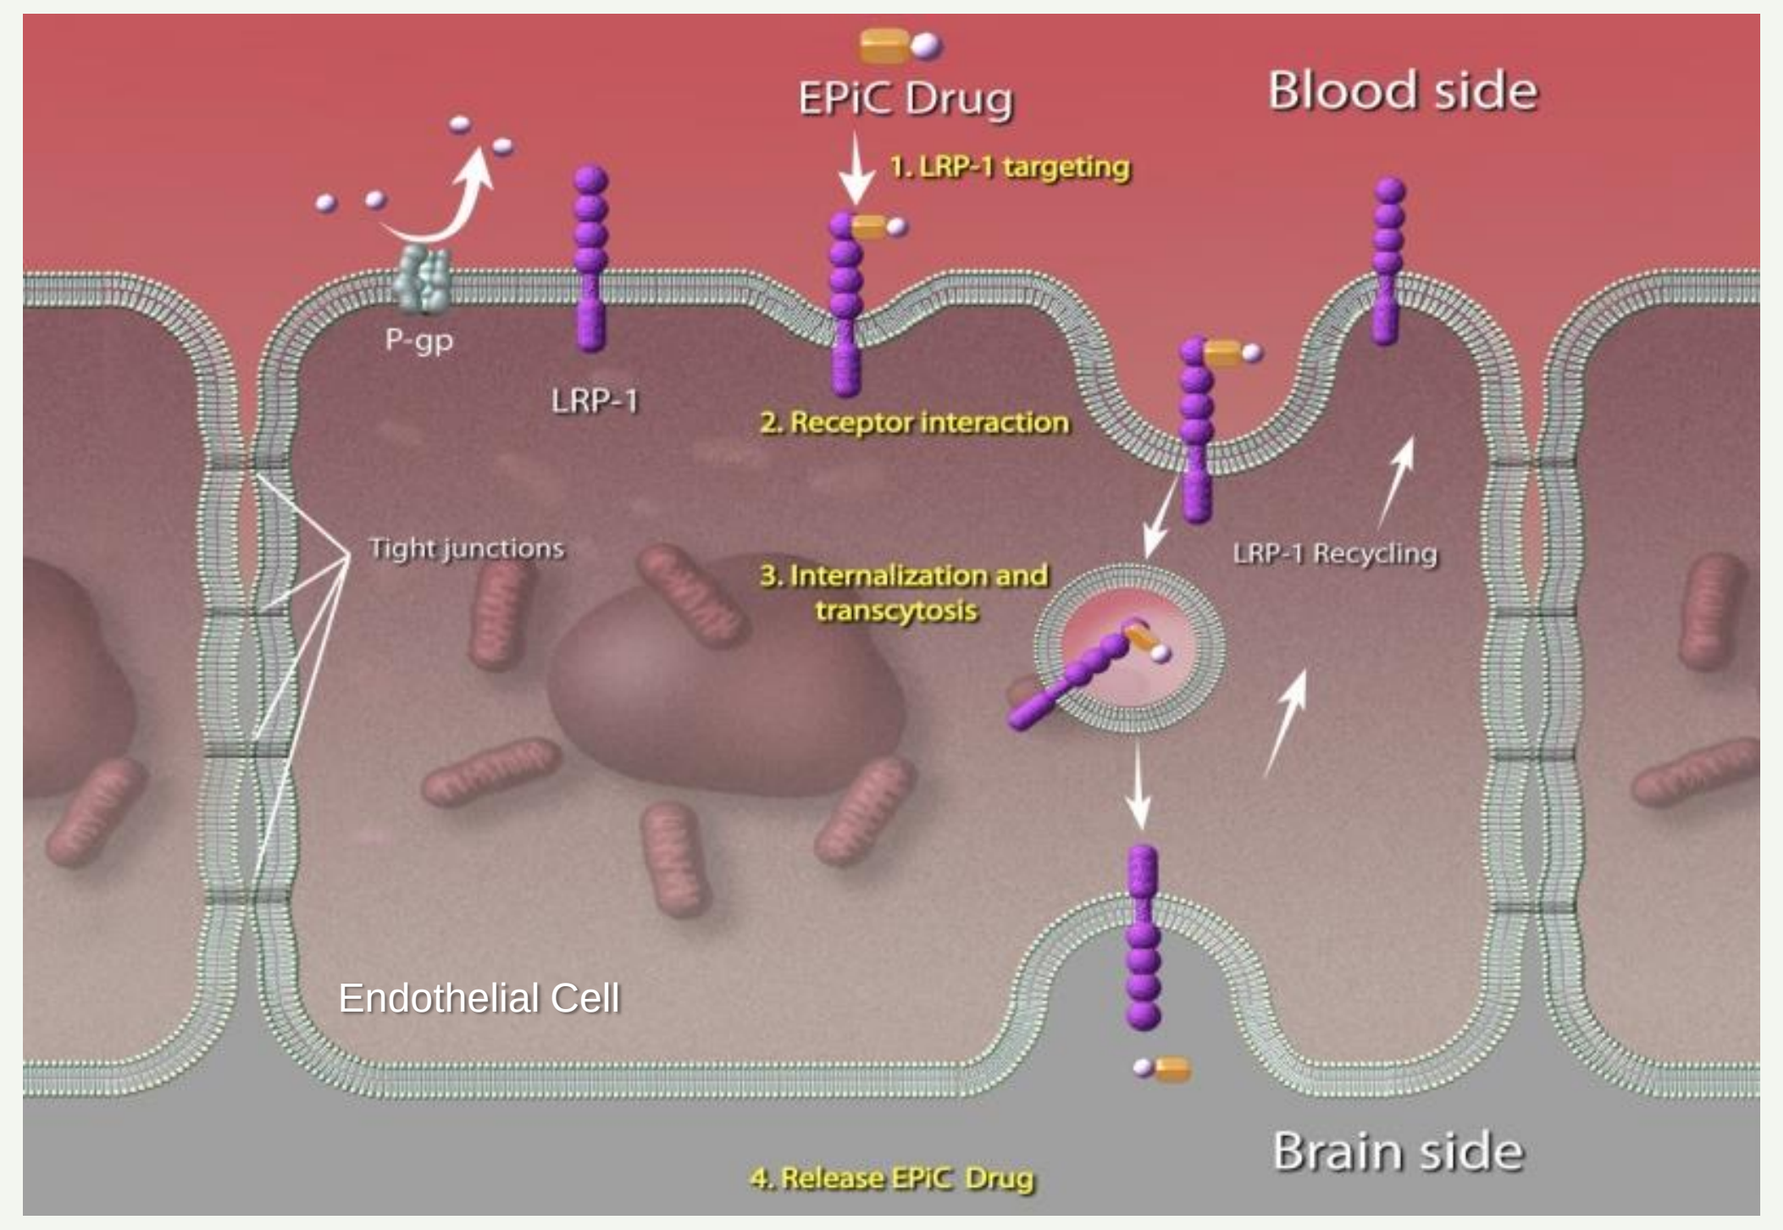
EPiC Drug
Blood side
1. LRP-1 targeting
P-gp
LRP-1
2. Receptor interaction
Tight junctions
3. Internalization and transcytosis
LRP-1 Recycling
Endothelial Cell
4. Release EPiC Drug
Brain side

Angiochem is a clinical-stage biotechnology company discovering and developing new breakthrough drugs that are uniquely capable of crossing the blood brain barrier (BBB) to treat brain diseases. These new drugs have the potential to address significant medical needs, many of which cannot be effectively addressed due to the fundamental physiological challenge the BBB presents.

Angiochem’s proprietary platform targets low-density lipoprotein receptor-related protein-1 (LRP1). This endogenous transcytosis system has a number of inherent biochemical advantages for drug transport across the BBB, including high capacity, rapid turnover, recognition of numerous ligands, and limited down-regulation. We have created peptides (Angiopeps), including Angiopep-2 (An2) using a library based on LPR-1 binding sequences of known LRP-1 ligands. These peptides can be introduced, by chemical conjugation or recombinant fusion, to small molecules and biologics, thus forming NCEs that are brain-penetrant Peptide-Drug Conjugates.

ANG4043

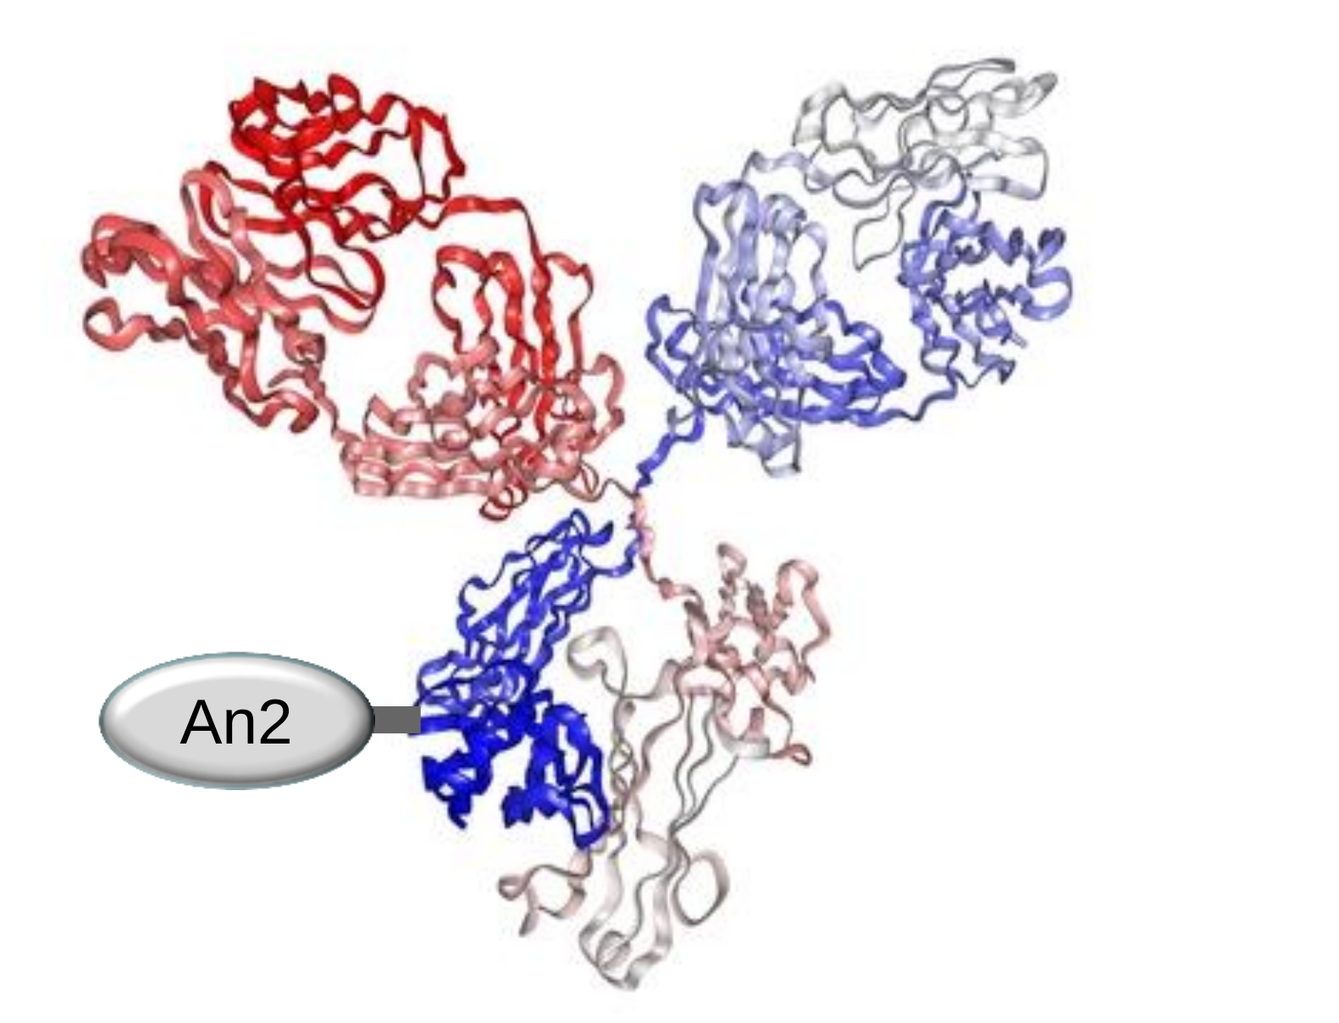
An2

- HER2 is the target of Herceptin (trastuzumab, Genentech)
- ANG4043 is a chemical conjugate of the anti-HER2 mAb with the peptide Angiopep-2 using a stable linker.
- ANG4043 is not a biosimilar, but a new chemical entiry.
- ANG4043 has potential to be a First-in-Class agent for HER2+ breast cancer brain metastases.
- The affinity of ANG4043 for the HER2 receptor is similar to that of the native (unconjugated) mAb.
- The anti-proliferative potency of ANG4043 is similar to that of the native mAb

ANG4043 Enters Brain and Targets Tumors

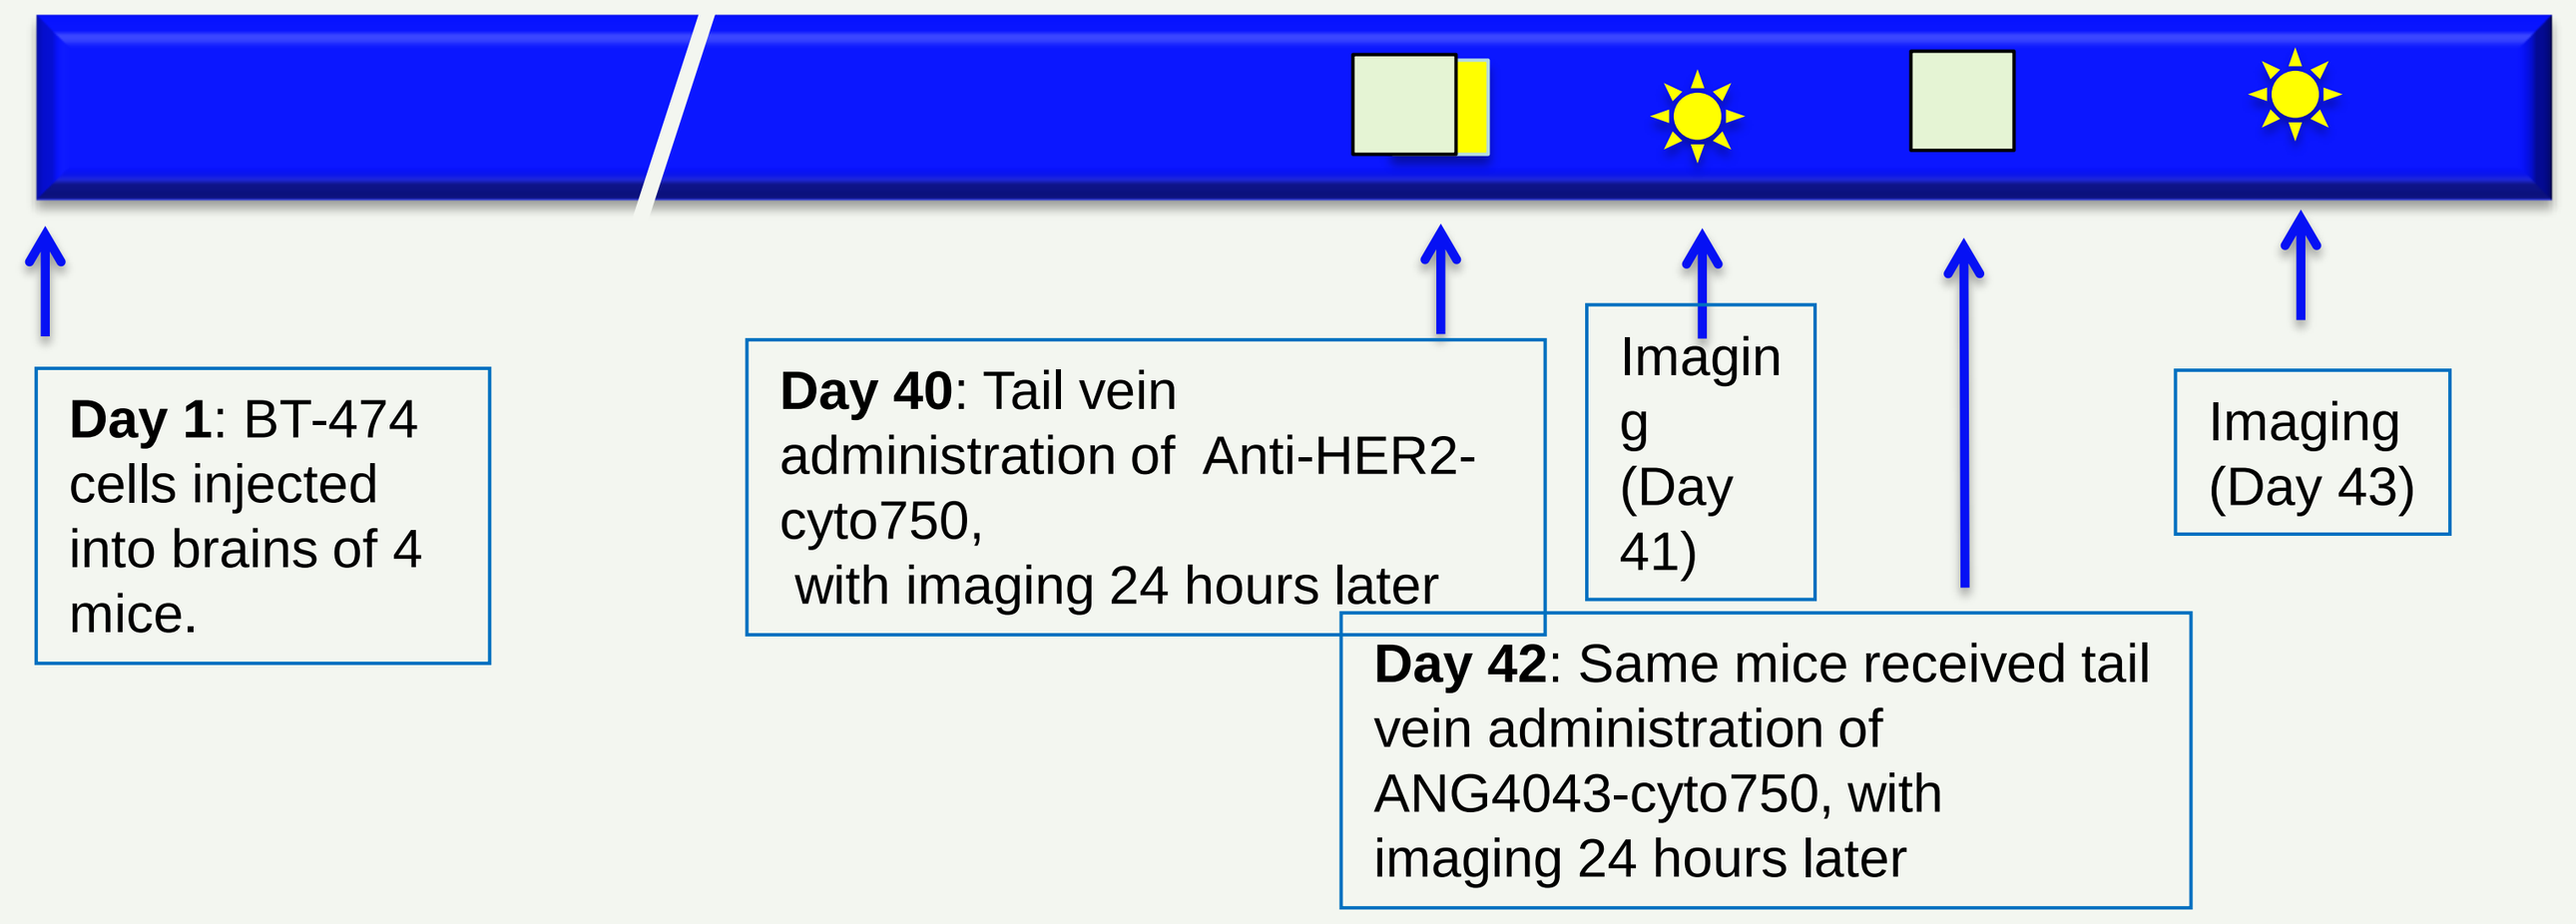
Day 1: BT-474 cells injected into brains of 4 mice.
Day 40: Tail vein administration of Anti-HER2-cyto750, with imaging 24 hours later
Imaging (Day 41)
Day 42: Same mice received tail vein administration of ANG4043-cyto750, with imaging 24 hours later
Imaging (Day 43)

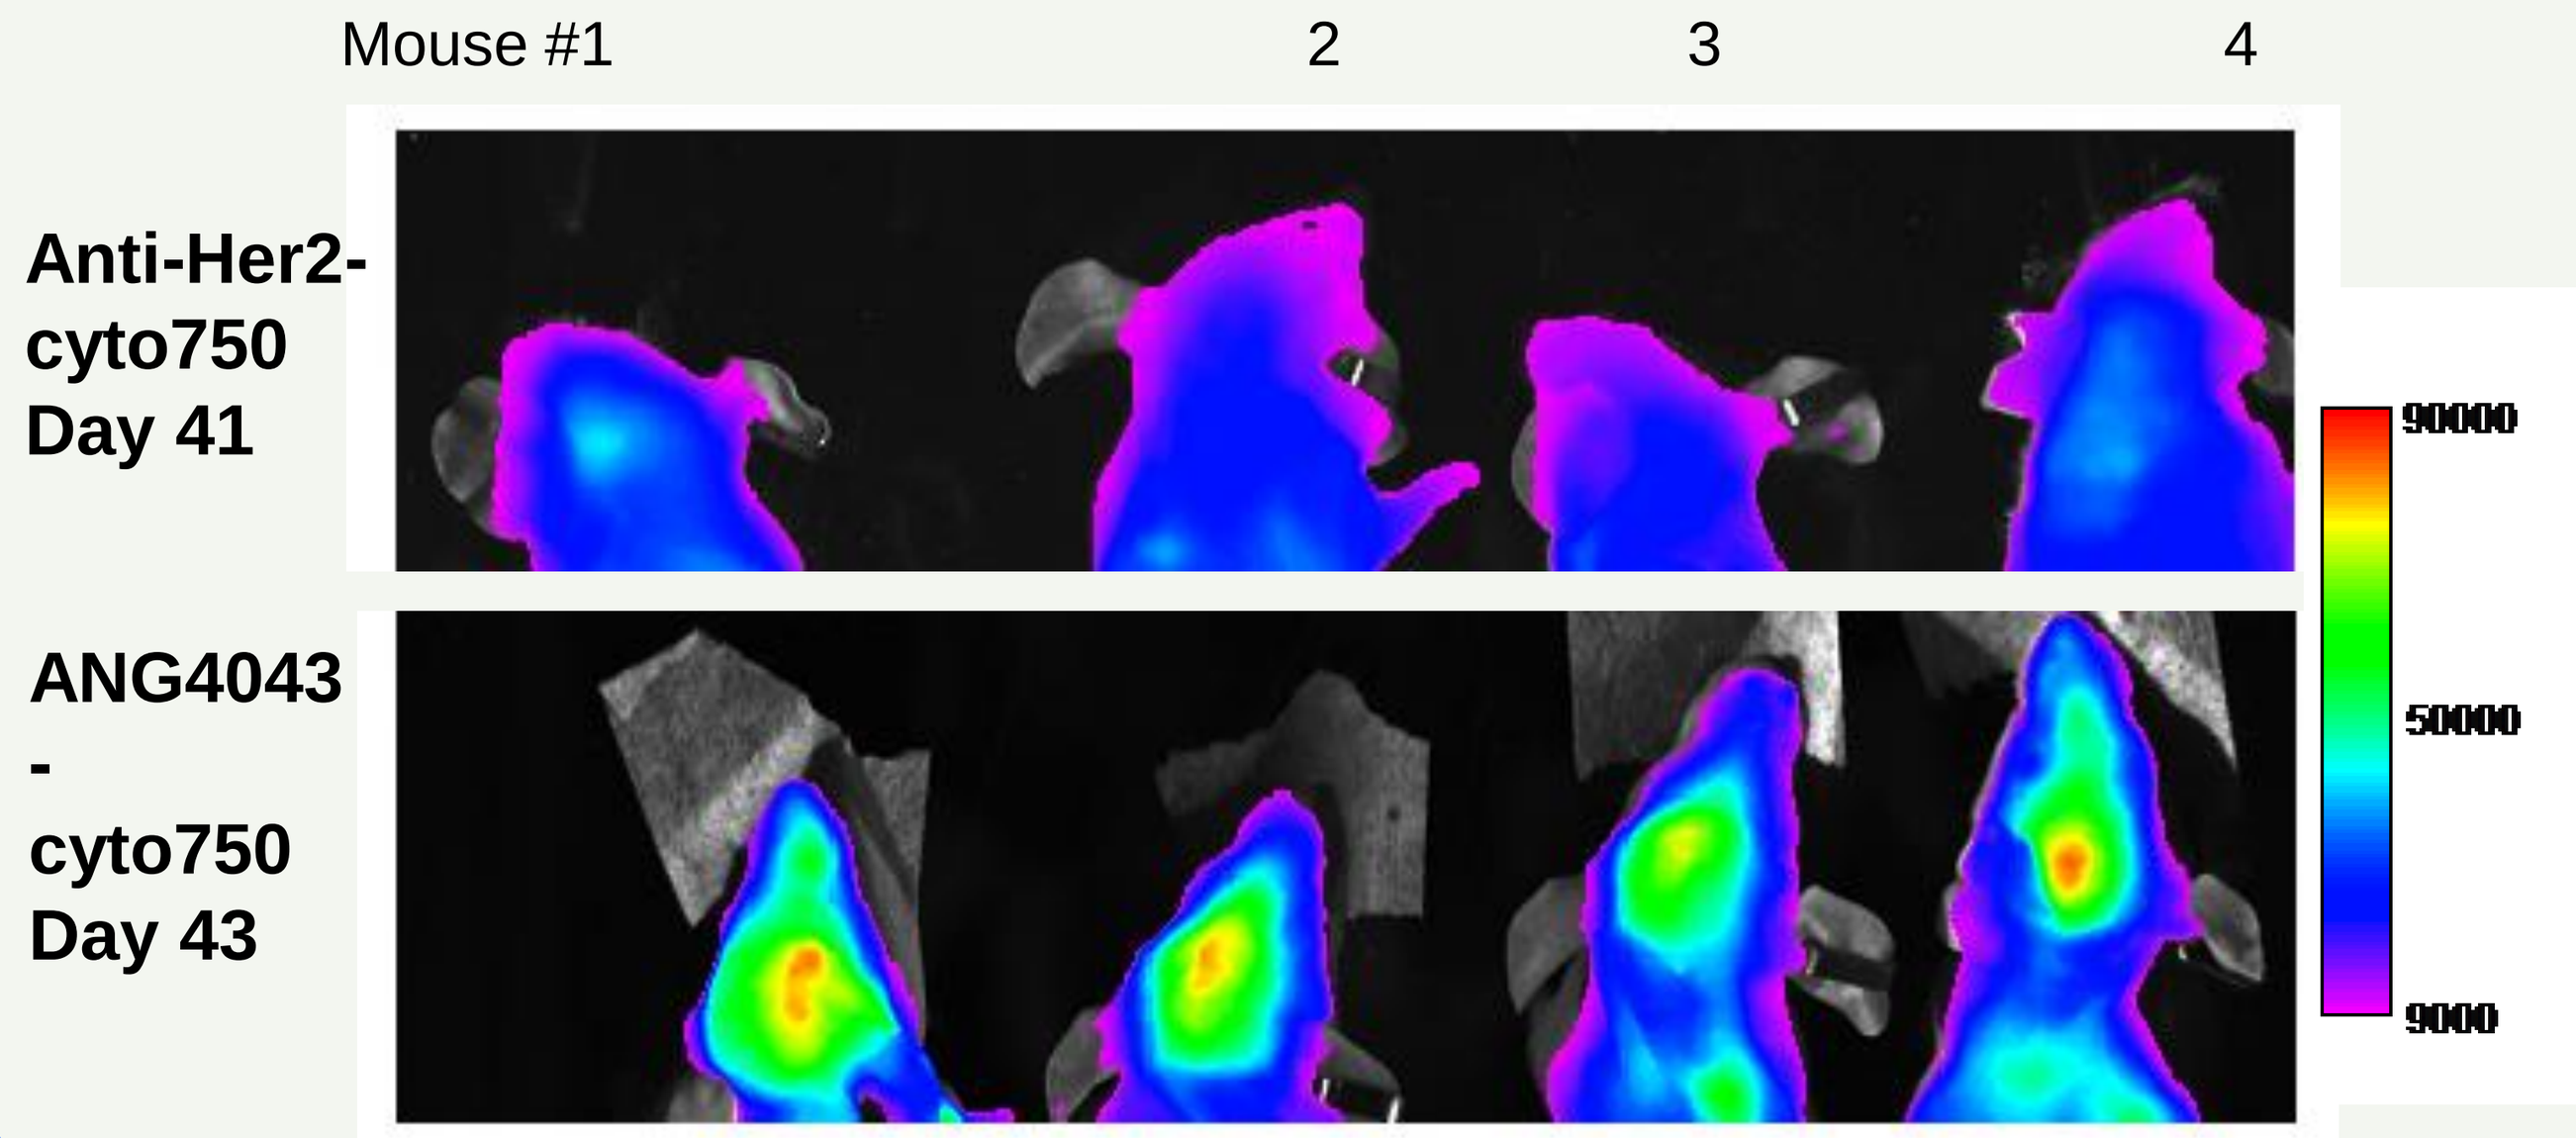
Mouse #1
2
3
4
Anti-Her2-cyto750 Day 41
ANG4043-cyto750 Day 43
90000
50000
9000

ANG4043 Reduces Tumor Mass

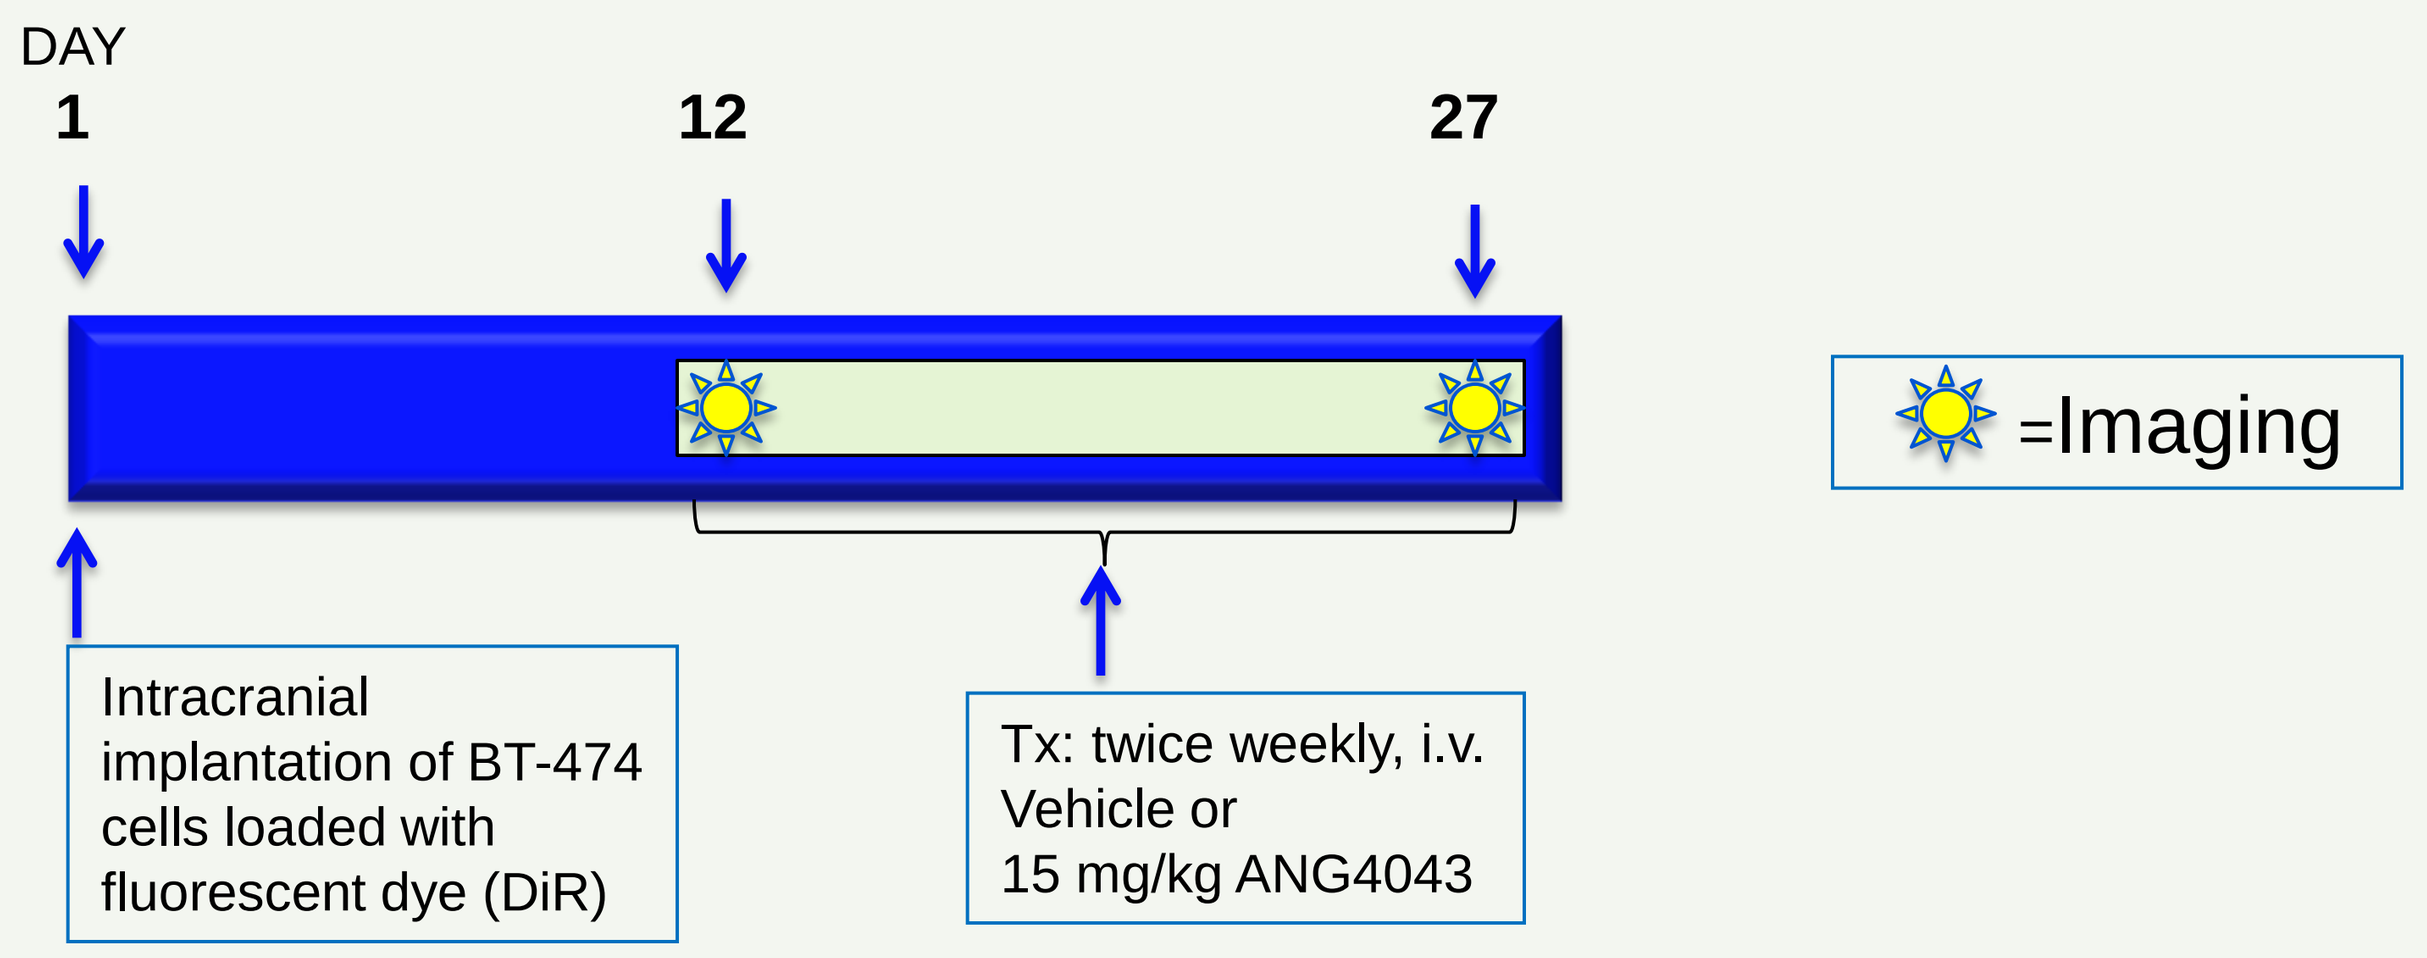
DAY
1
12
27
=Imaging
Intracranial implantation of BT-474 cells loaded with fluorescent dye (DiR)
Tx: twice weekly, i.v. Vehicle or 15 mg/kg ANG4043

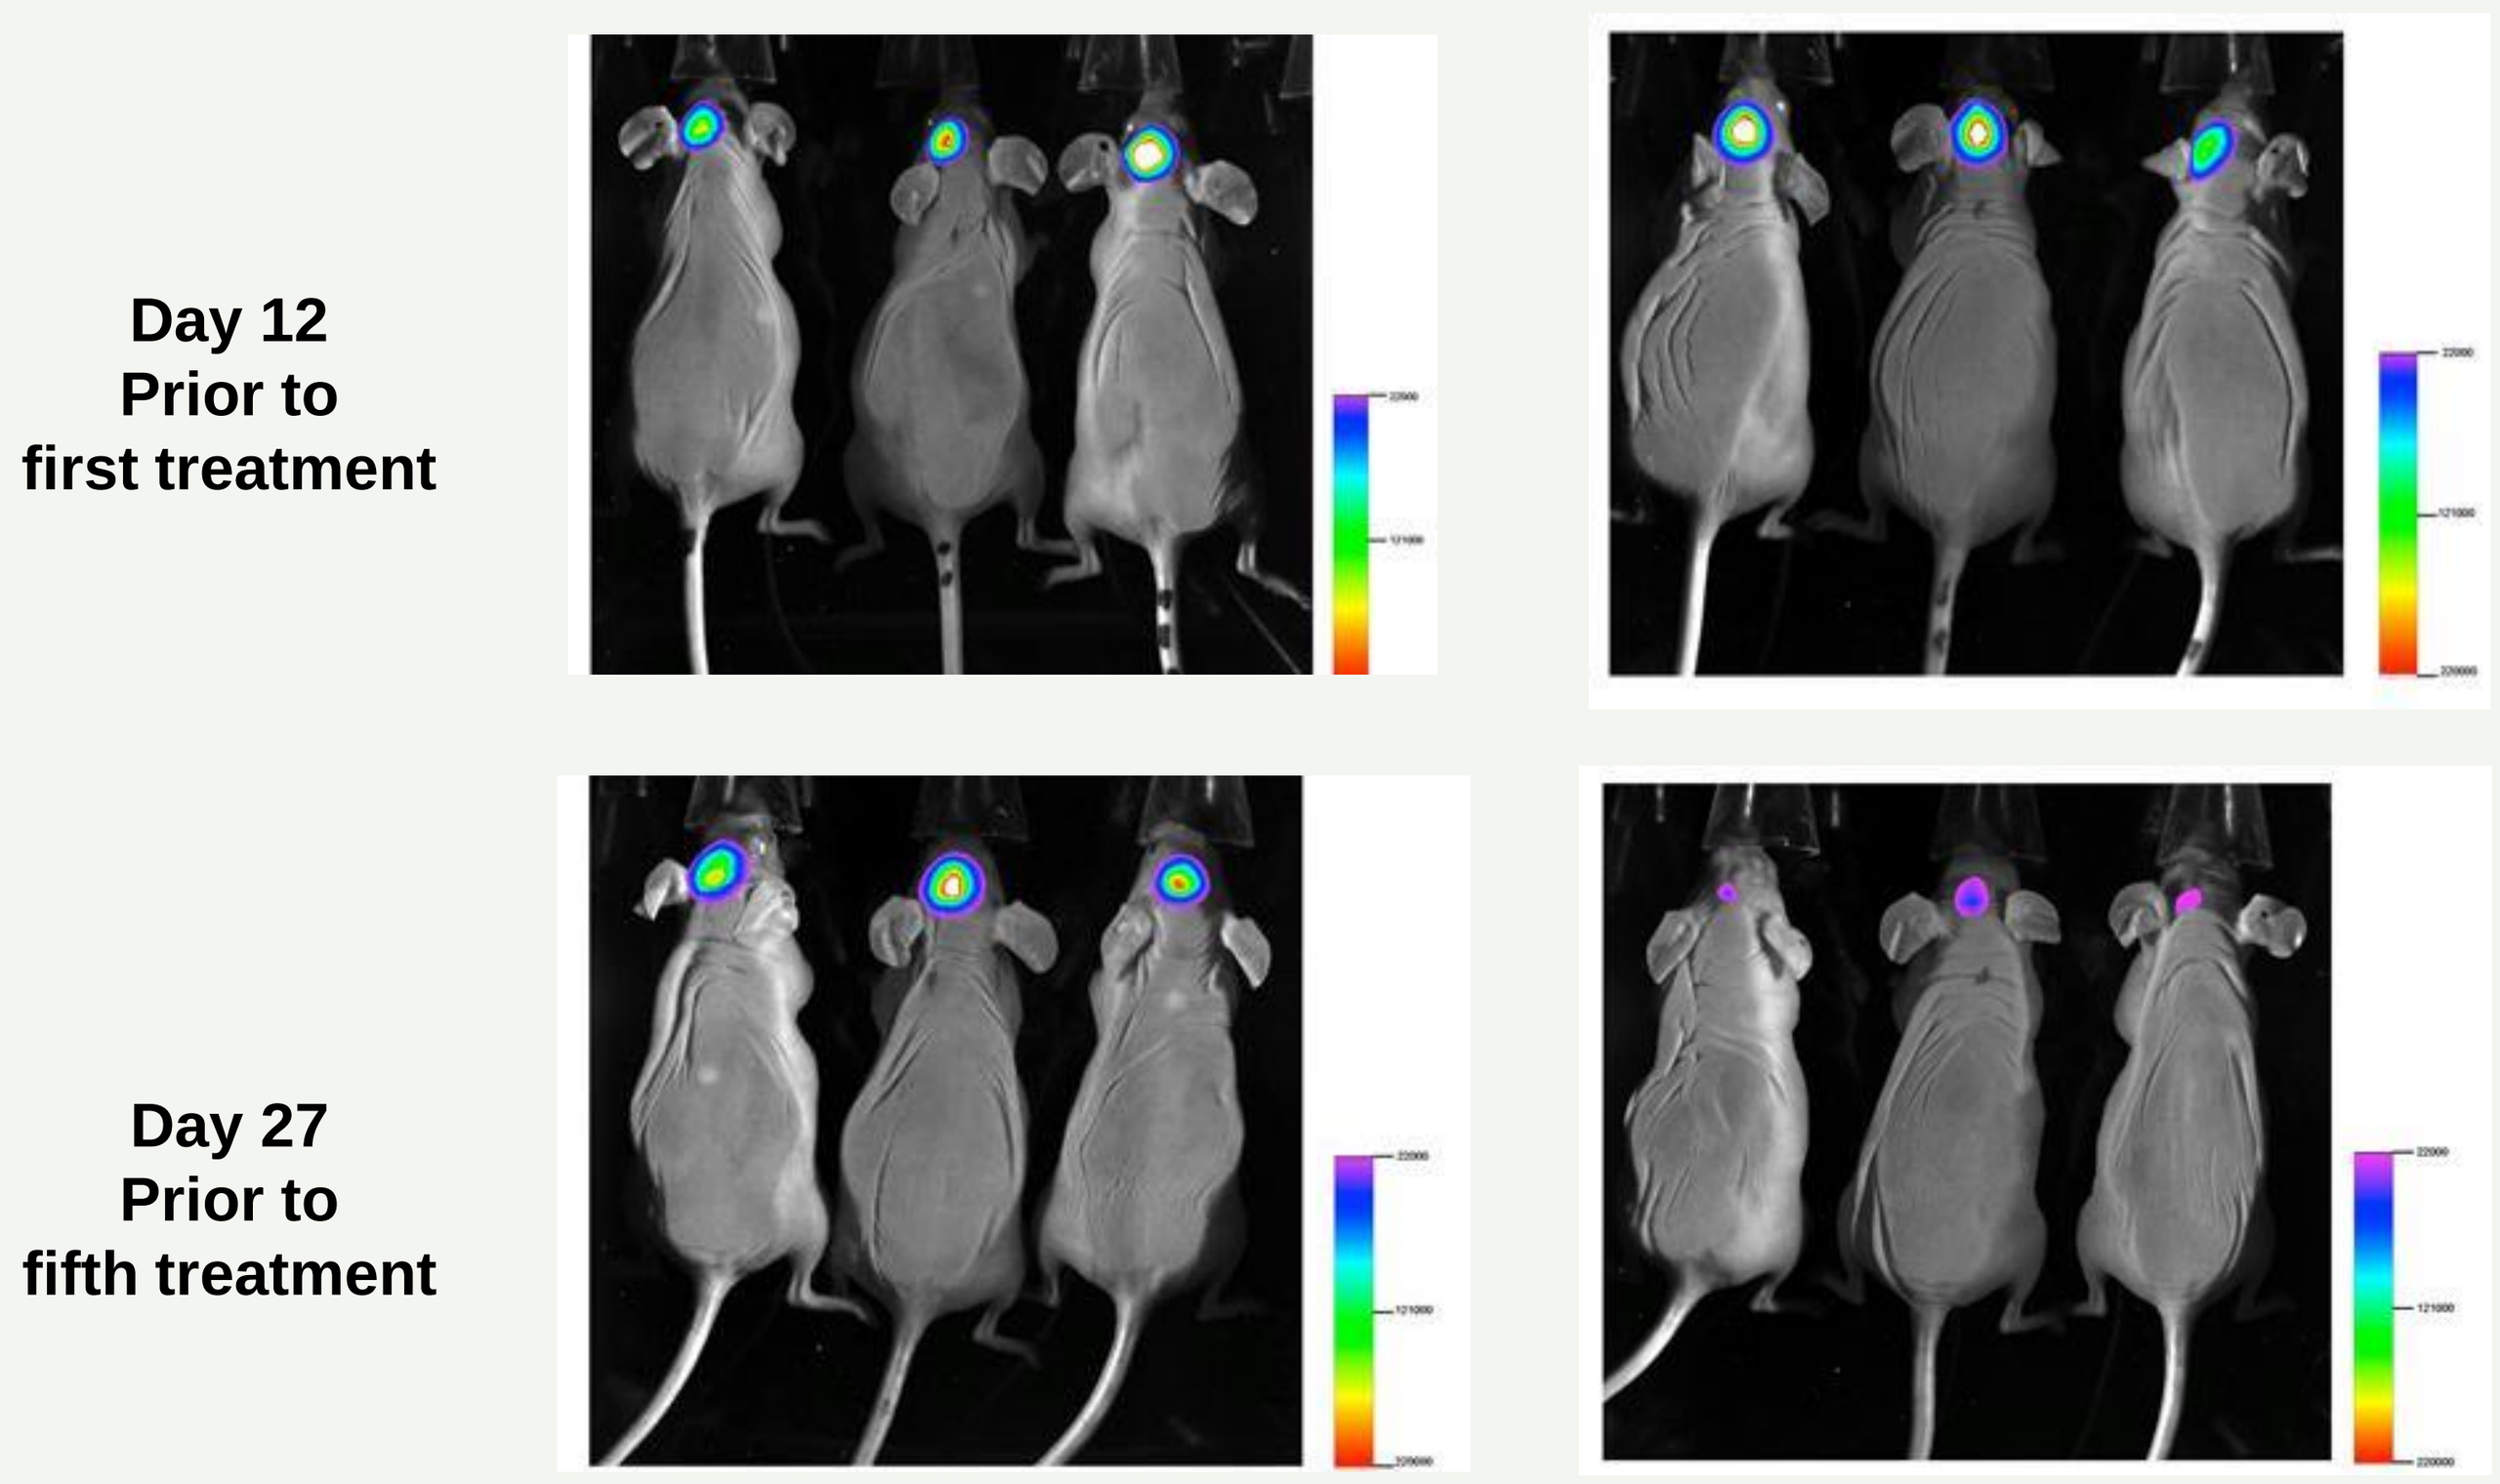
Day 12 Prior to first treatment
Day 27 Prior to fifth treatment

ANG4043 Increases Survival

- Day 1: Intracranial implantation of BT-474 tumor cells to mice
- Day 12: Treatment begins: twice weekly, i.v.
- Body weight and morbidity/mortality monitored
- Comparison study (n=10), shown in figure below.
 - Vehicle
 - Anti-HER2 mAb (5 mg/kg)
 - ANG4043 (5 mg/kg equivalent)
- Dose Response Study
 - Vehicle
 - ANG4043 (5 mg/kg)
 - ANG4043 (15 mg/kg)

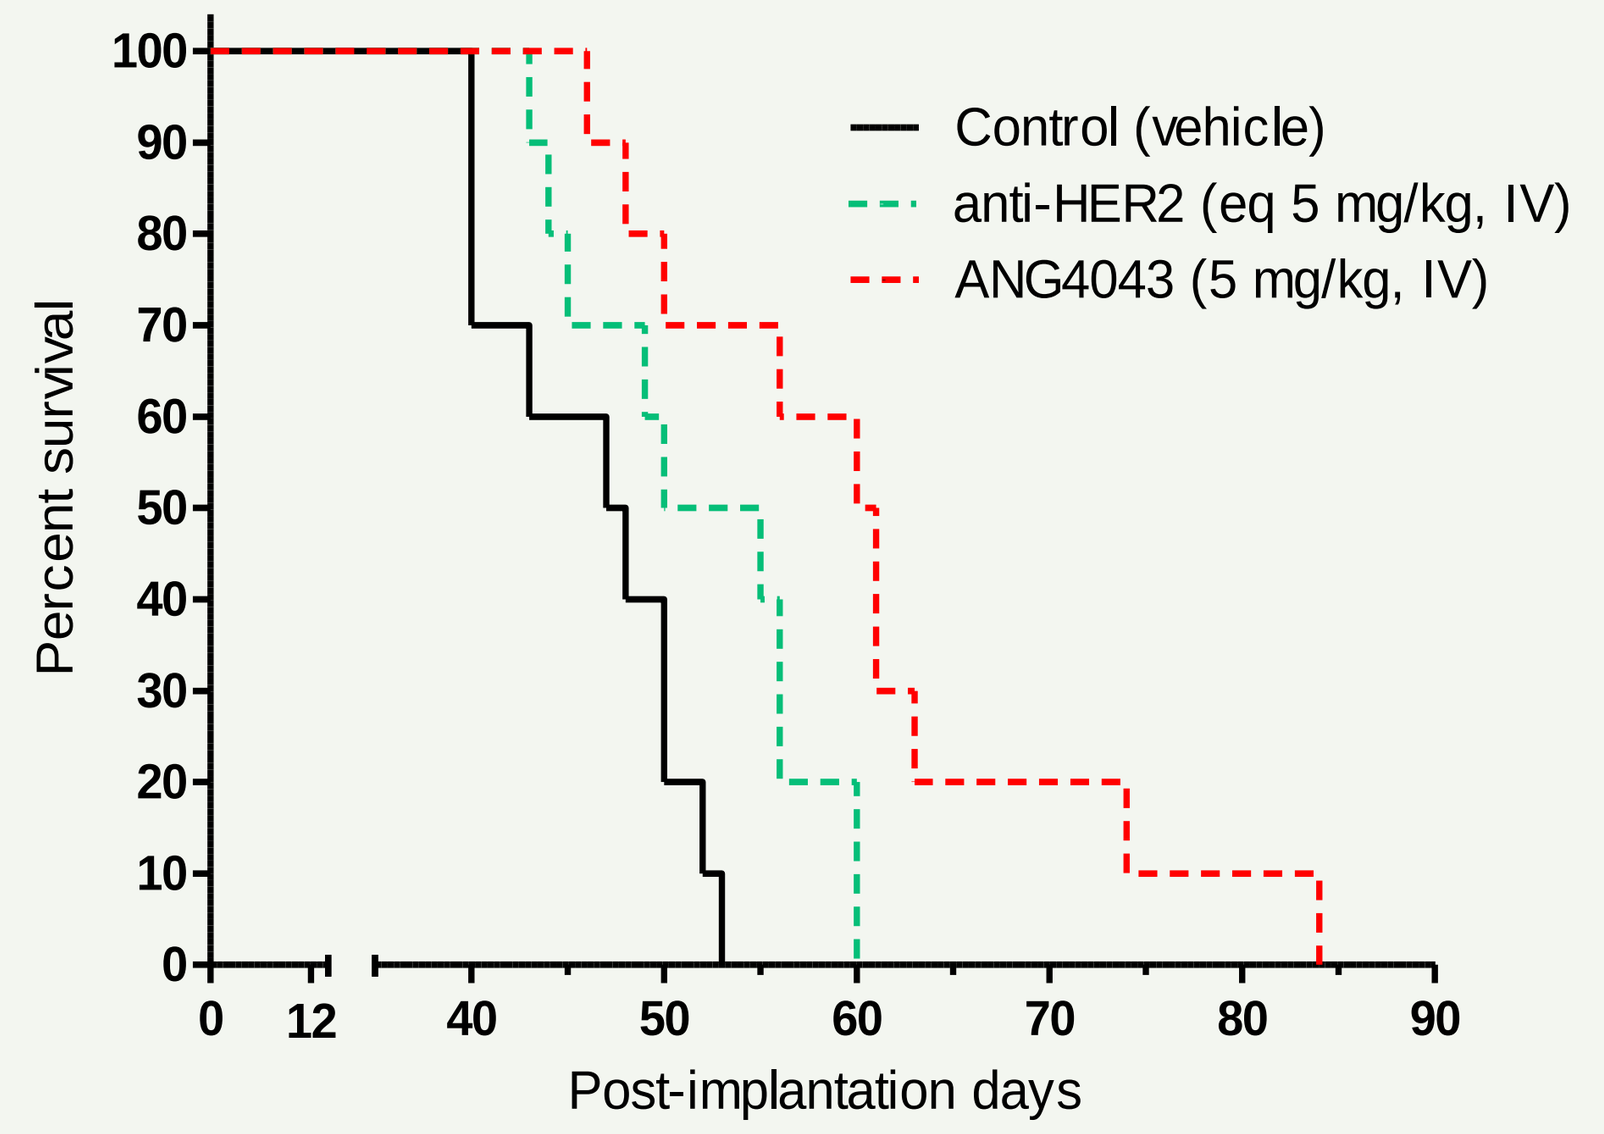
Percent survival
Post-implantation days
Control (vehicle)
anti-HER2 (eq 5 mg/kg, IV)
ANG4043 (5 mg/kg, IV)

Survival Study Summary

	Control	Herceptin (5 mg/kg)	ANG4043 (5 mg/kg)	ANG4043 (15 mg/kg)
Mean	46 days	52 days	64 days	80 days
% increase vs. control		+13%	+39%	+74%
P value		p=0.028	p=0.0012	p=0.0003

Conclusions

- **The brain-penetrant peptide-mAb conjugate, ANG4043, targets intracranial tumors and shrinks tumor size in mice, indicating that therapeutic concentrations have been achieved in brain.**
- **This reduction in tumor size translates to a significant increase in survival in mice with intracranial HER2+ (BT-474 cell) tumors.**
- **Mice treated with ANG4043, while displaying improved survival, do eventually die, suggesting that a mechanism for resistance to anti-HER2 therapy emerges in the tumor cells.**
- **As current clinical guidelines for Herceptin treatment include co-administration with a cytotoxic drug for peripheral HER2+ primary and metastatic tumors, HER2+ brain metastases will likely require co-administration of a brain penetrant mAb (ANG4043) and a brain-penetrant cytotoxic drug, such as the An2-paclitaxel conjugate ANG1005.**